A CLINICAL EVALUATION OF A GEL WITH BETA-GLUCAN*
IN THE MANAGEMENT OF NON-INFECTED DIABETIC FOOT ULCERATIONS

Donna Welch, *Advanced Podiatrist - Diabetes, Humber NHS Foundation Trust*

Introduction

Managing diabetic foot ulcerations can be a challenge due to the various complications that can affect the healing process (Basu and Shukla, 2012).

Unlike other chronic wounds, progression may be delayed by altered neutrophil function, diminished tissue perfusion and/or defective protein synthesis (Wounds International, 2013).

In addition, plantar pressures during weight bearing activity in conjunction with peripheral neuropathy may lead to continued trauma.

Prompt healing of any wound helps to reduce financial costs but more importantly minimises the physical and psychological effect to the individual patient. Clinicians have to consider the most appropriate dressing to optimise wound healing.

The product tested is a gel with Beta-glucan, consisting of water, glycerol, carboxymethylcellulose and Soluble Beta-glucan (SBG). SBG is a yeast derived compound that acts on cells to accelerate healing.

Method:

Patients were selected to the evaluation meeting the following criteria:

- **Wound measurements between 1 – 30cm^2**
- **Patient has a stalled wound for at least 4 weeks**
- **Wound exudate levels assessed as low to moderate**
- **Wound assessed as being free from classical signs of infection**
- **Patients HbA1c < 86mmol/mol**
- **Patient was able to give informed consent and be monitored throughout the evaluation period**

Patients consented to use of a gel with Beta-glucan as a primary dressing. All secondary dressings were consistent with previous treatments, Atrauman and secondary foam. Dressings were changed according to clinical need, on average 2-3 times weekly.

Appropriate sharp debridement and offloading was maintained throughout the evaluation period. Digital images were taken on a weekly basis.

Data collected included: wound dimensions, wound bed presentation, exudate levels and pain analogue score. The ease of application and response to treatment was also documented.

A CLINICAL EVALUATION OF A GEL WITH BETA-GLUCAN*
IN THE MANAGEMENT OF NON-INFECTED DIABETIC FOOT ULCERATIONS

Case Study 1:

Female patient aged 54 years had left leg bypass graft and amputation of left 3-4 toes, following critical limb ischaemia. Newly diagnosed Diabetic with a HbA1c 64 mmol/mol.

Patient presented with a left foot dorsal ulceration of 4 week duration. Ulcer measured 20mm x 25mm with adherent slough and necrosis to base, and minimal granulation tissue to margins.

Patient reported a pain scale of 3 and agreed to trial the gel with Beta-glucan. After 2 weeks of application, the dorsal wound had improved with reduced slough and necrosis. Pain scale was now recorded as 0 and the patient was very satisfied with the results.

At week 2, the patient requested the gel with Beta-glucan be used on her amputation site also, as current dressing was adhering and causing discomfort.

ULCER #1

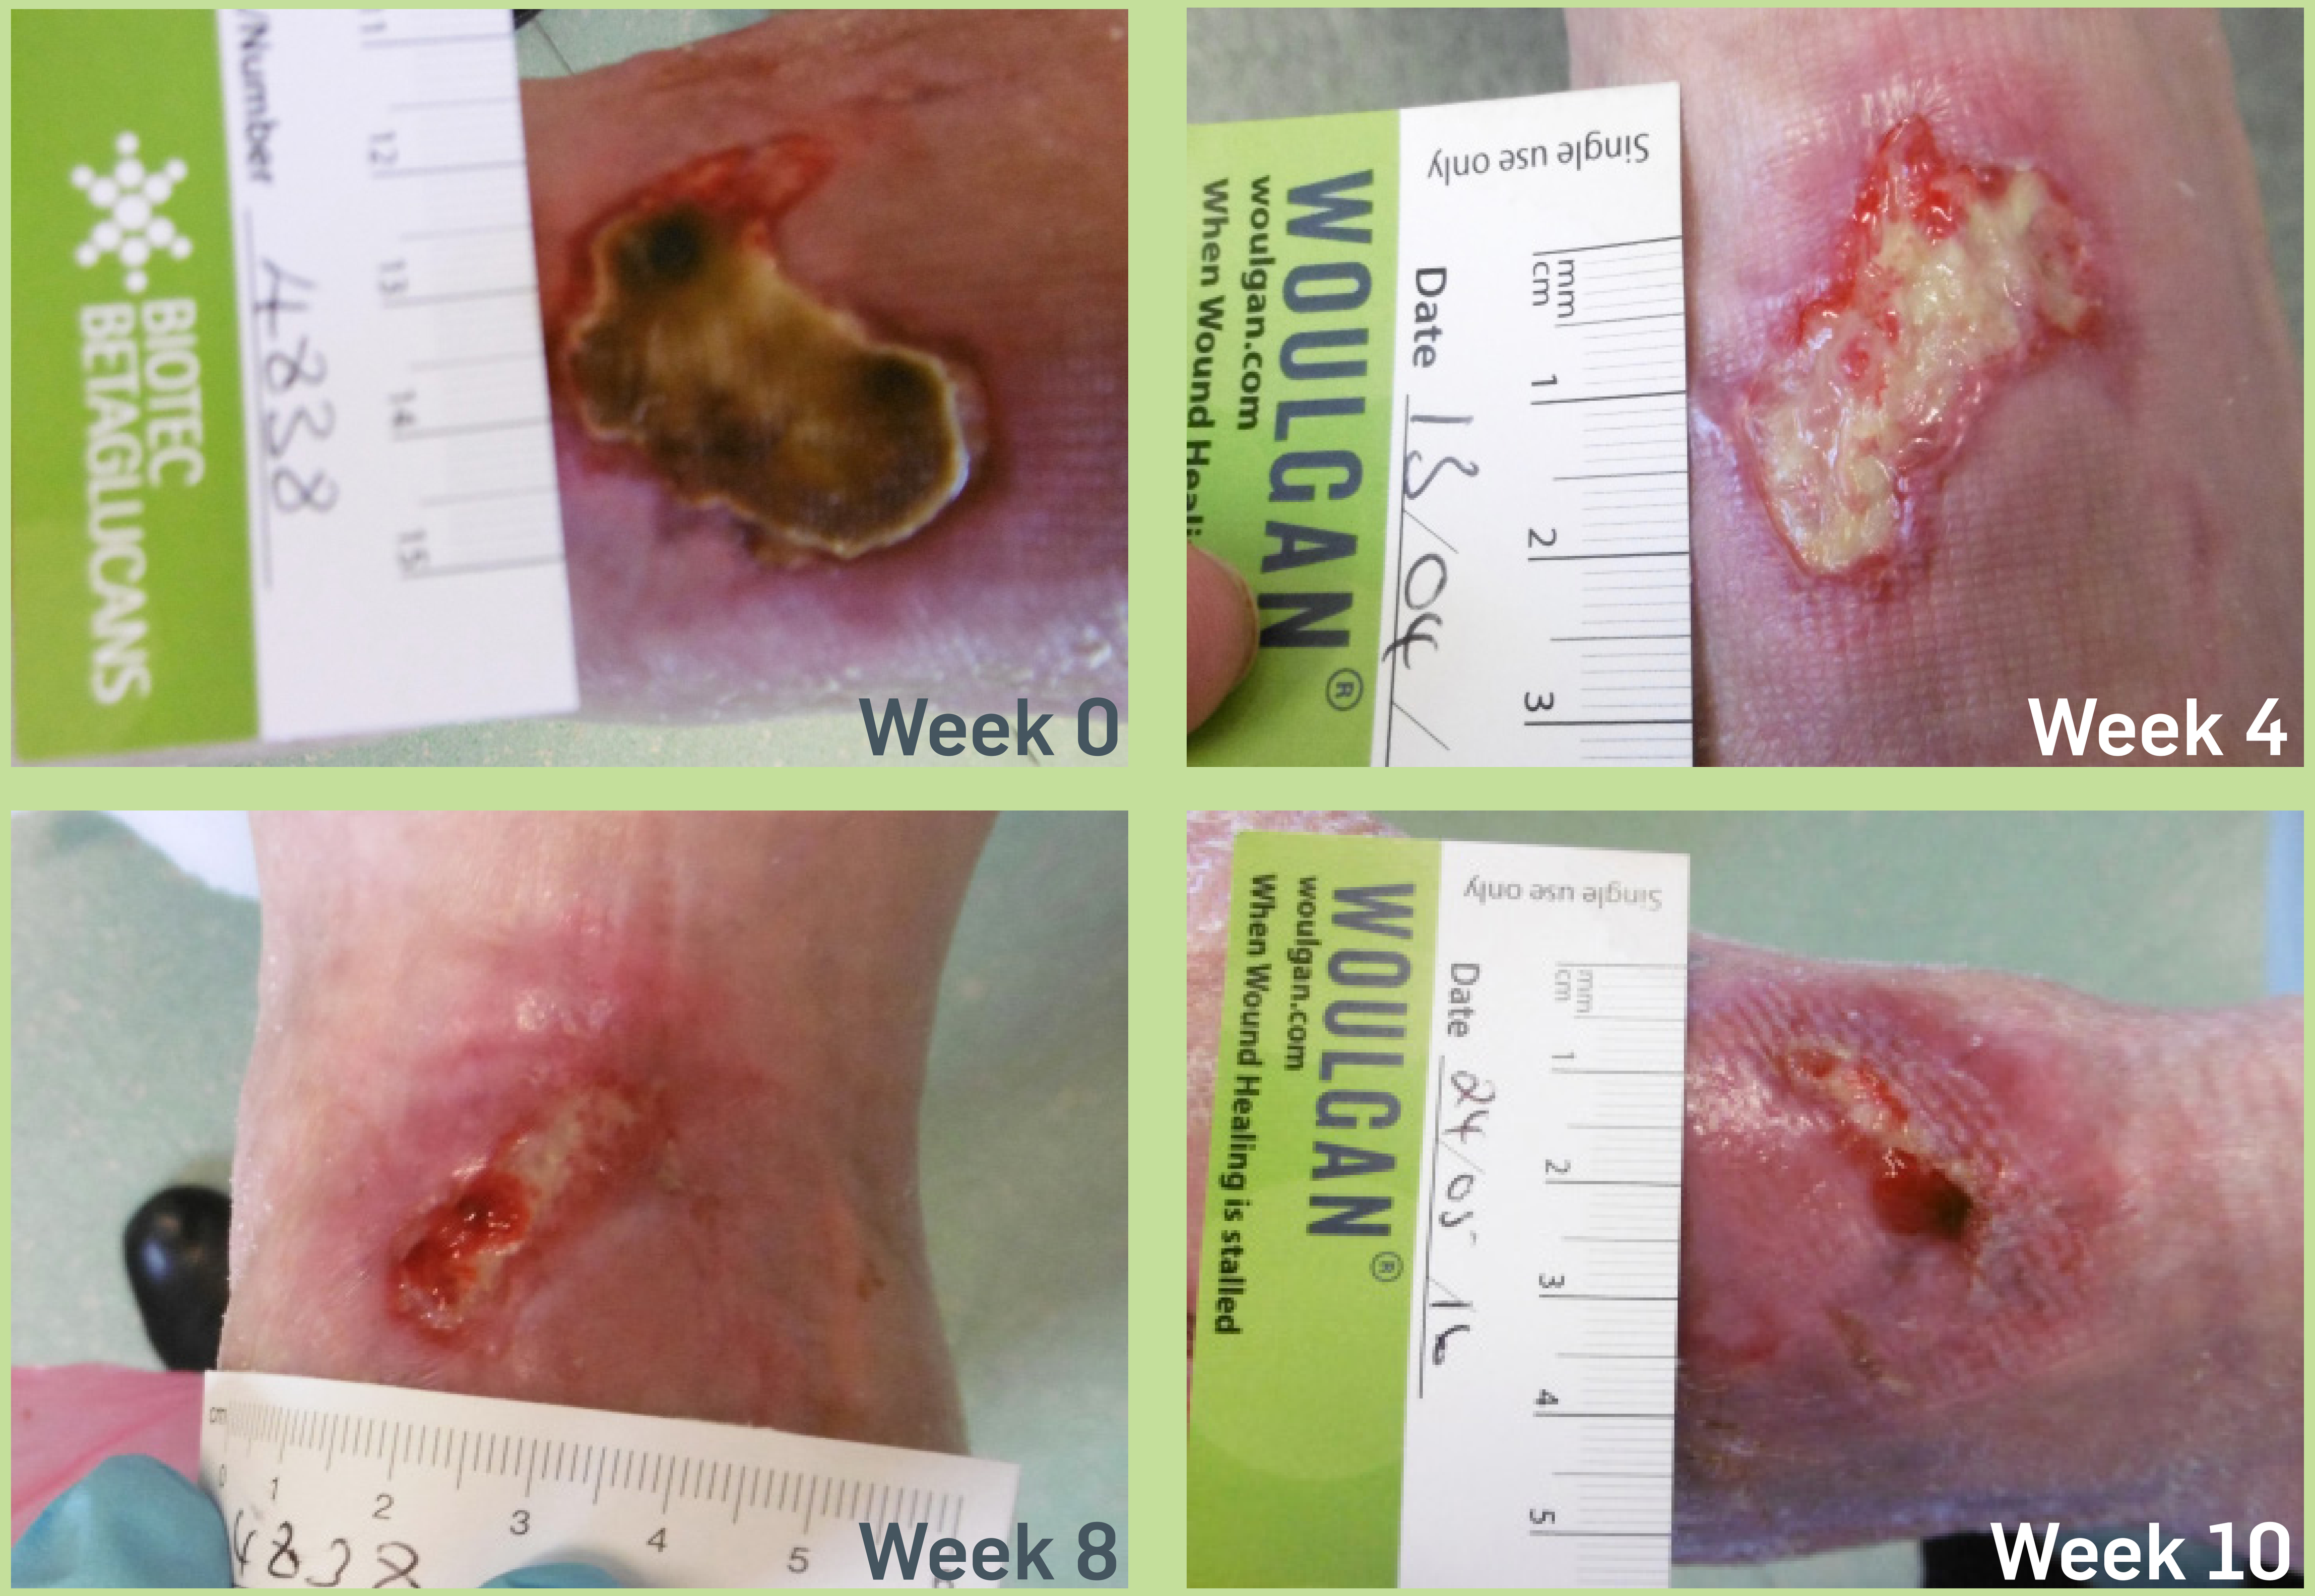
BIOTEC BETAGLUCANS
Number 4838
Week 0
WOULGAN
woulgan.com
Single use only
Date 13/04
Week 4
Week 8
WOULGAN
woulgan.com
When Wound Healing is stalled
Single use only
Date 24/05/16
Week 10

ULCER #2

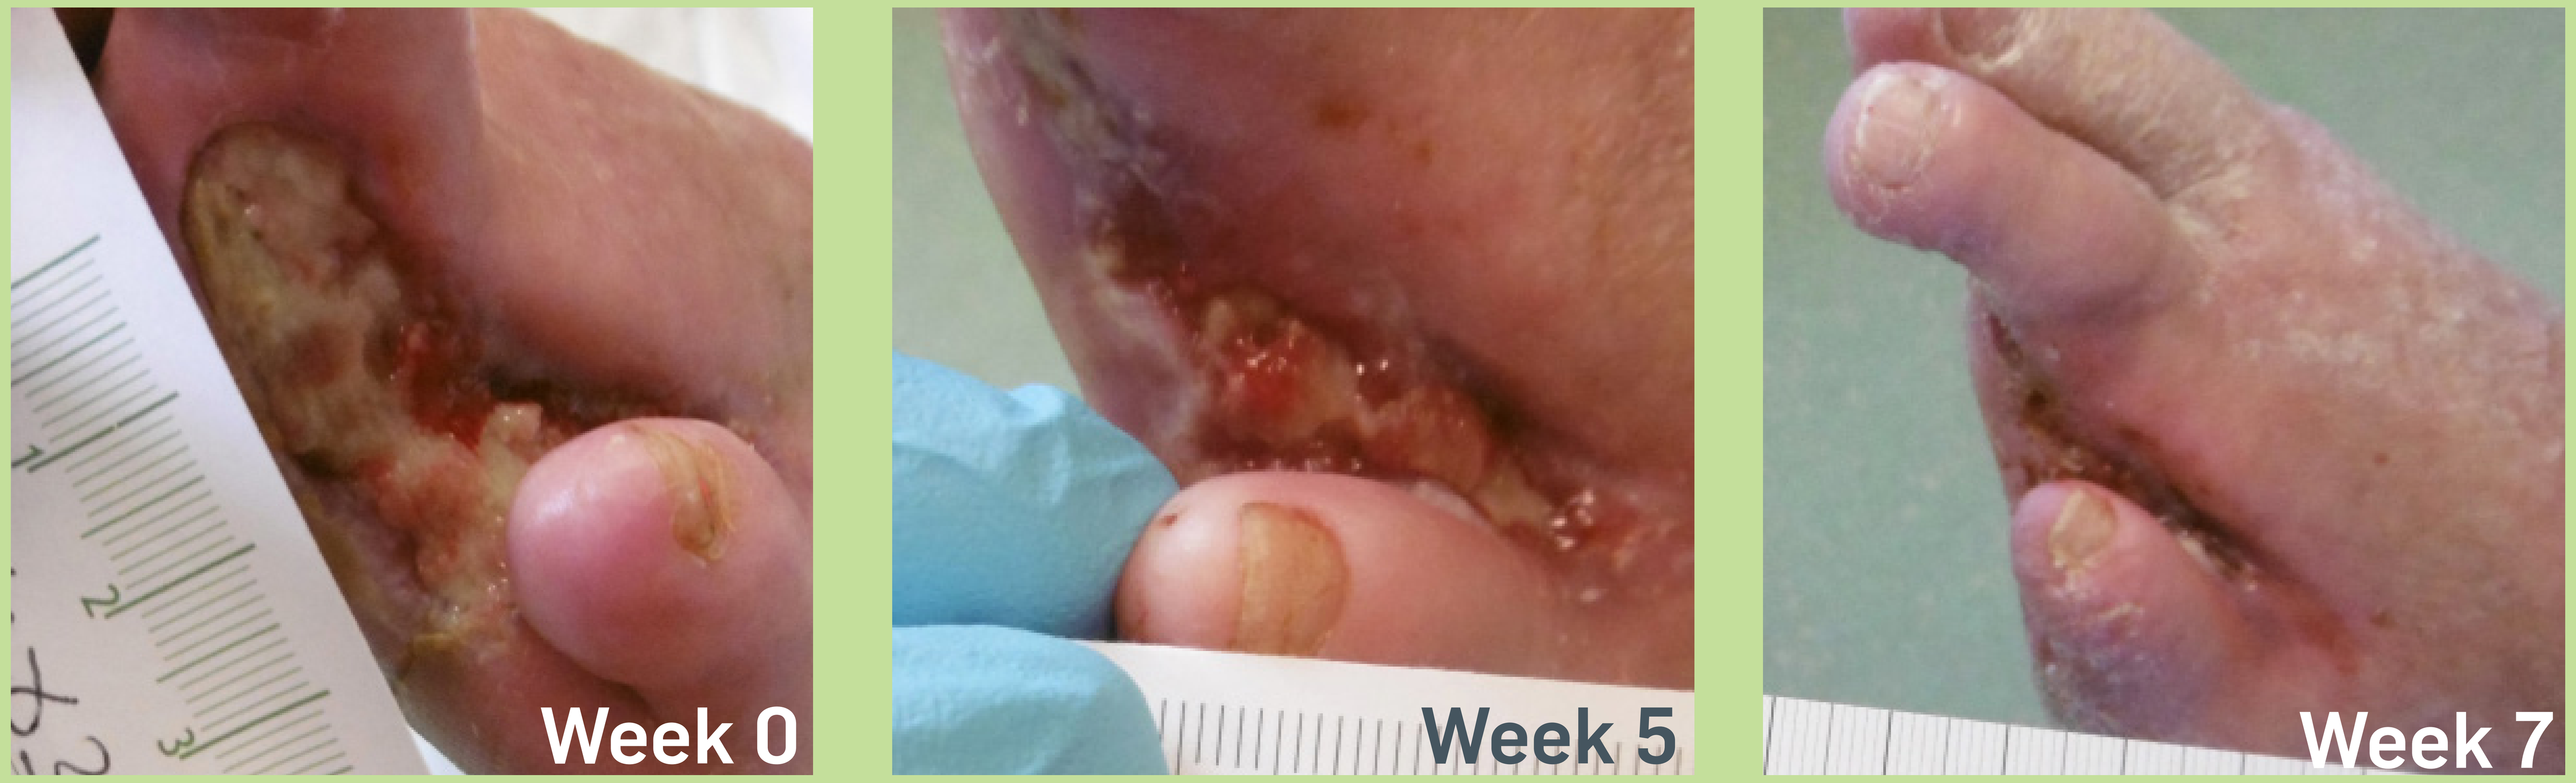
Week 0
Week 5
Week 7

The amputation site was assessed at this stage as being suitable for use of the gel containing Beta-glucan. The dressing increased patient comfort and caused minimal trauma to new tissue.

Both ulcerations continued to improve with less slough visible in wound bed and healthy tissue forming. Both wounds decreased in size at each review and completely healed within 12 weeks.

A CLINICAL EVALUATION OF A GEL WITH BETA-GLUCAN*
IN THE MANAGEMENT OF NON-INFECTED DIABETIC FOOT ULCERATIONS

Case Study 2:

Male patient aged 49 years old. HbA1c 67mmol/mol. Right forefoot amputation 2010, which had never completely healed. Previous management has included various dressing types, bespoke footwear and filler. Patient required 1–2 weekly appointments within the Diabetic Foot Clinic and additional weekly wound clinic appointments with community nurses for dressings. Patient had suffered repeated episodes of infection requiring antibiotic treatment.

Wound was assessed as appropriate for use of a gel with Beta-glucan, measuring 5mm x 10mm with low exudate levels, no pain and no signs of infection. In week 1, the patient presented with minimal change to the wound bed, but surrounding tissue and wound margins looked healthier.

The ulcer continued to progress until it was completely healed at week 9. Patient remains intact 10 months after stopping the trial.

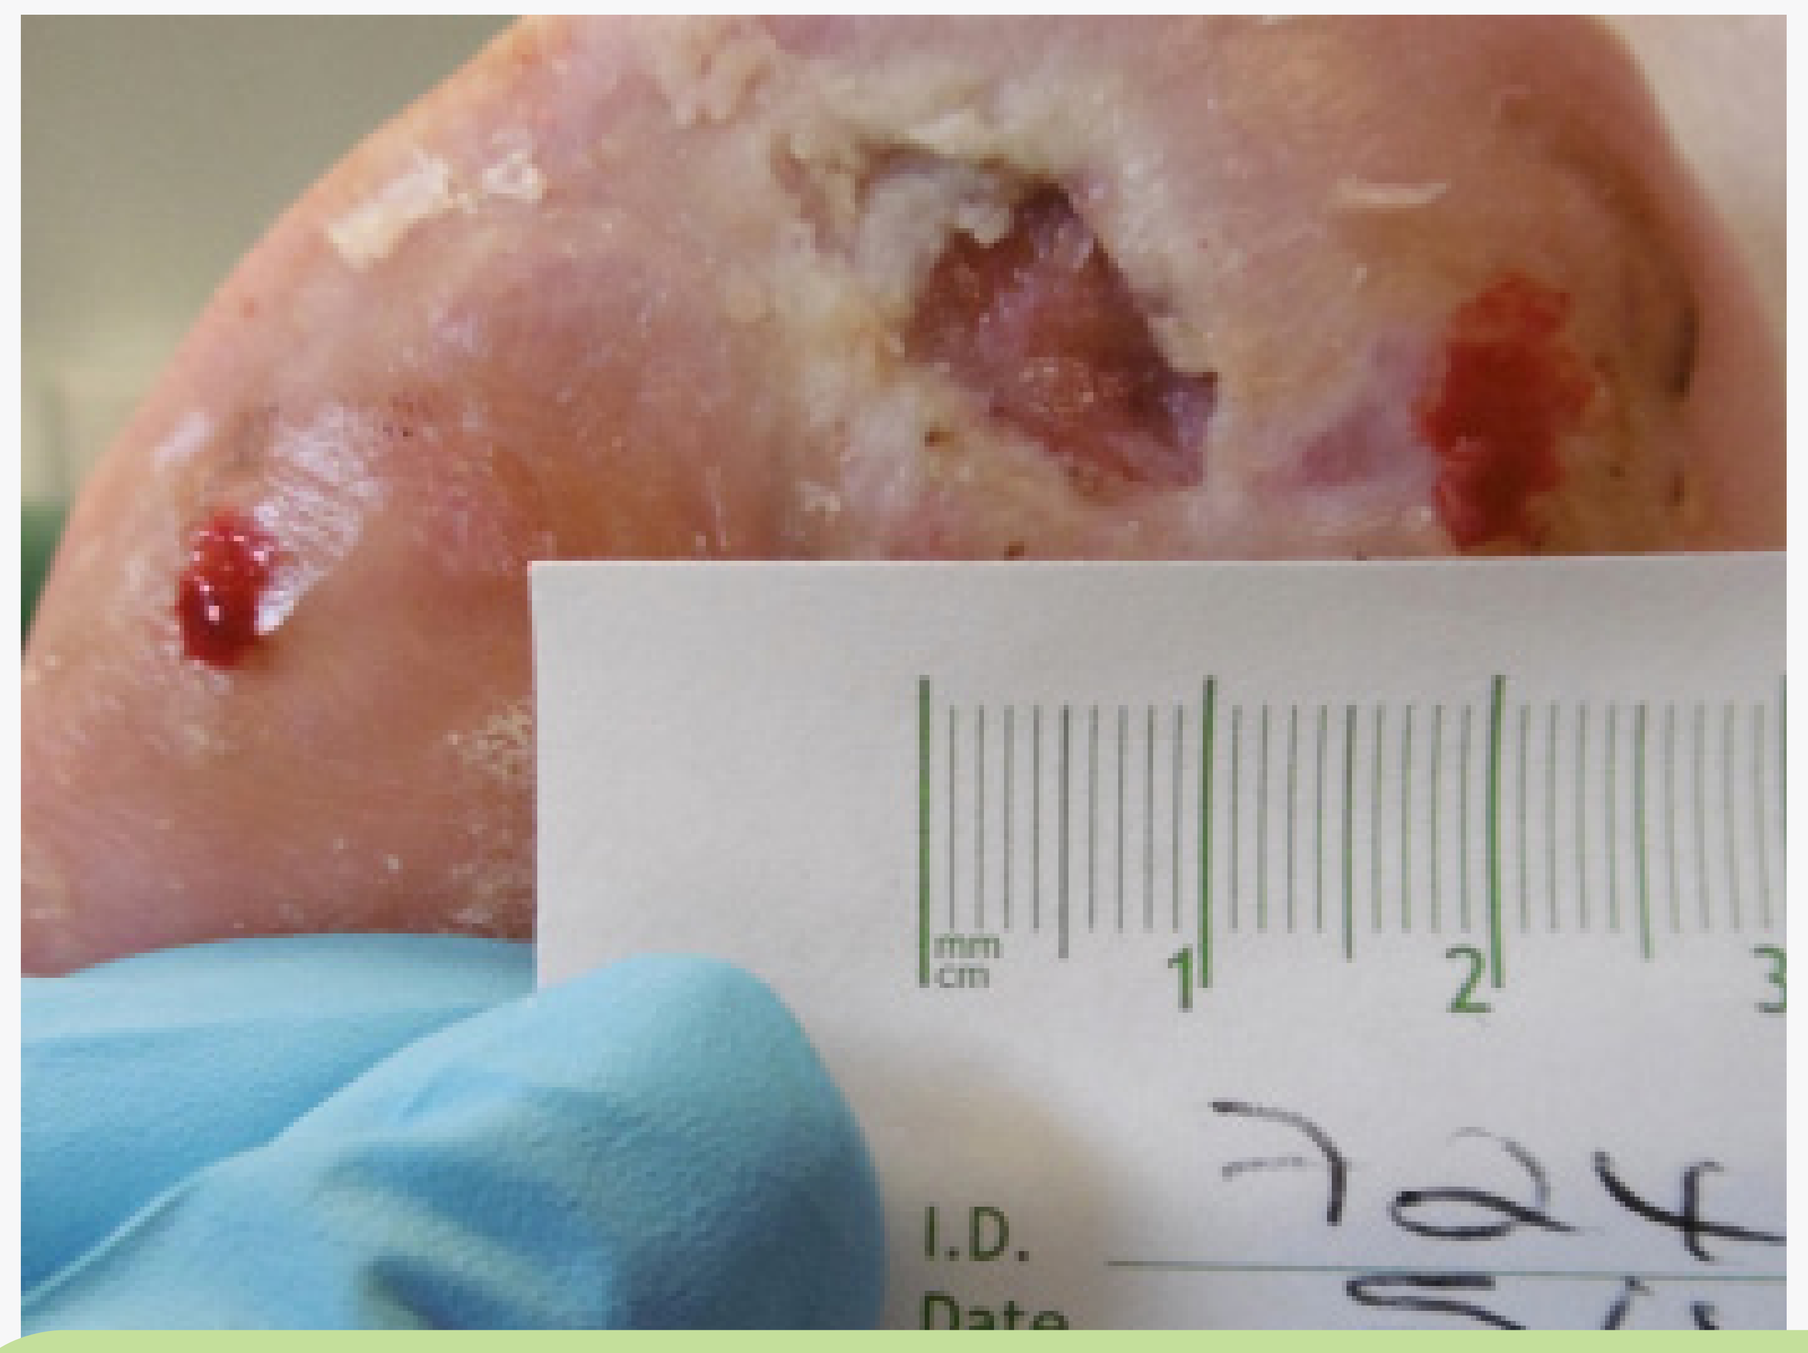
mm
cm
1
2
3
I.D.
724

Week 1

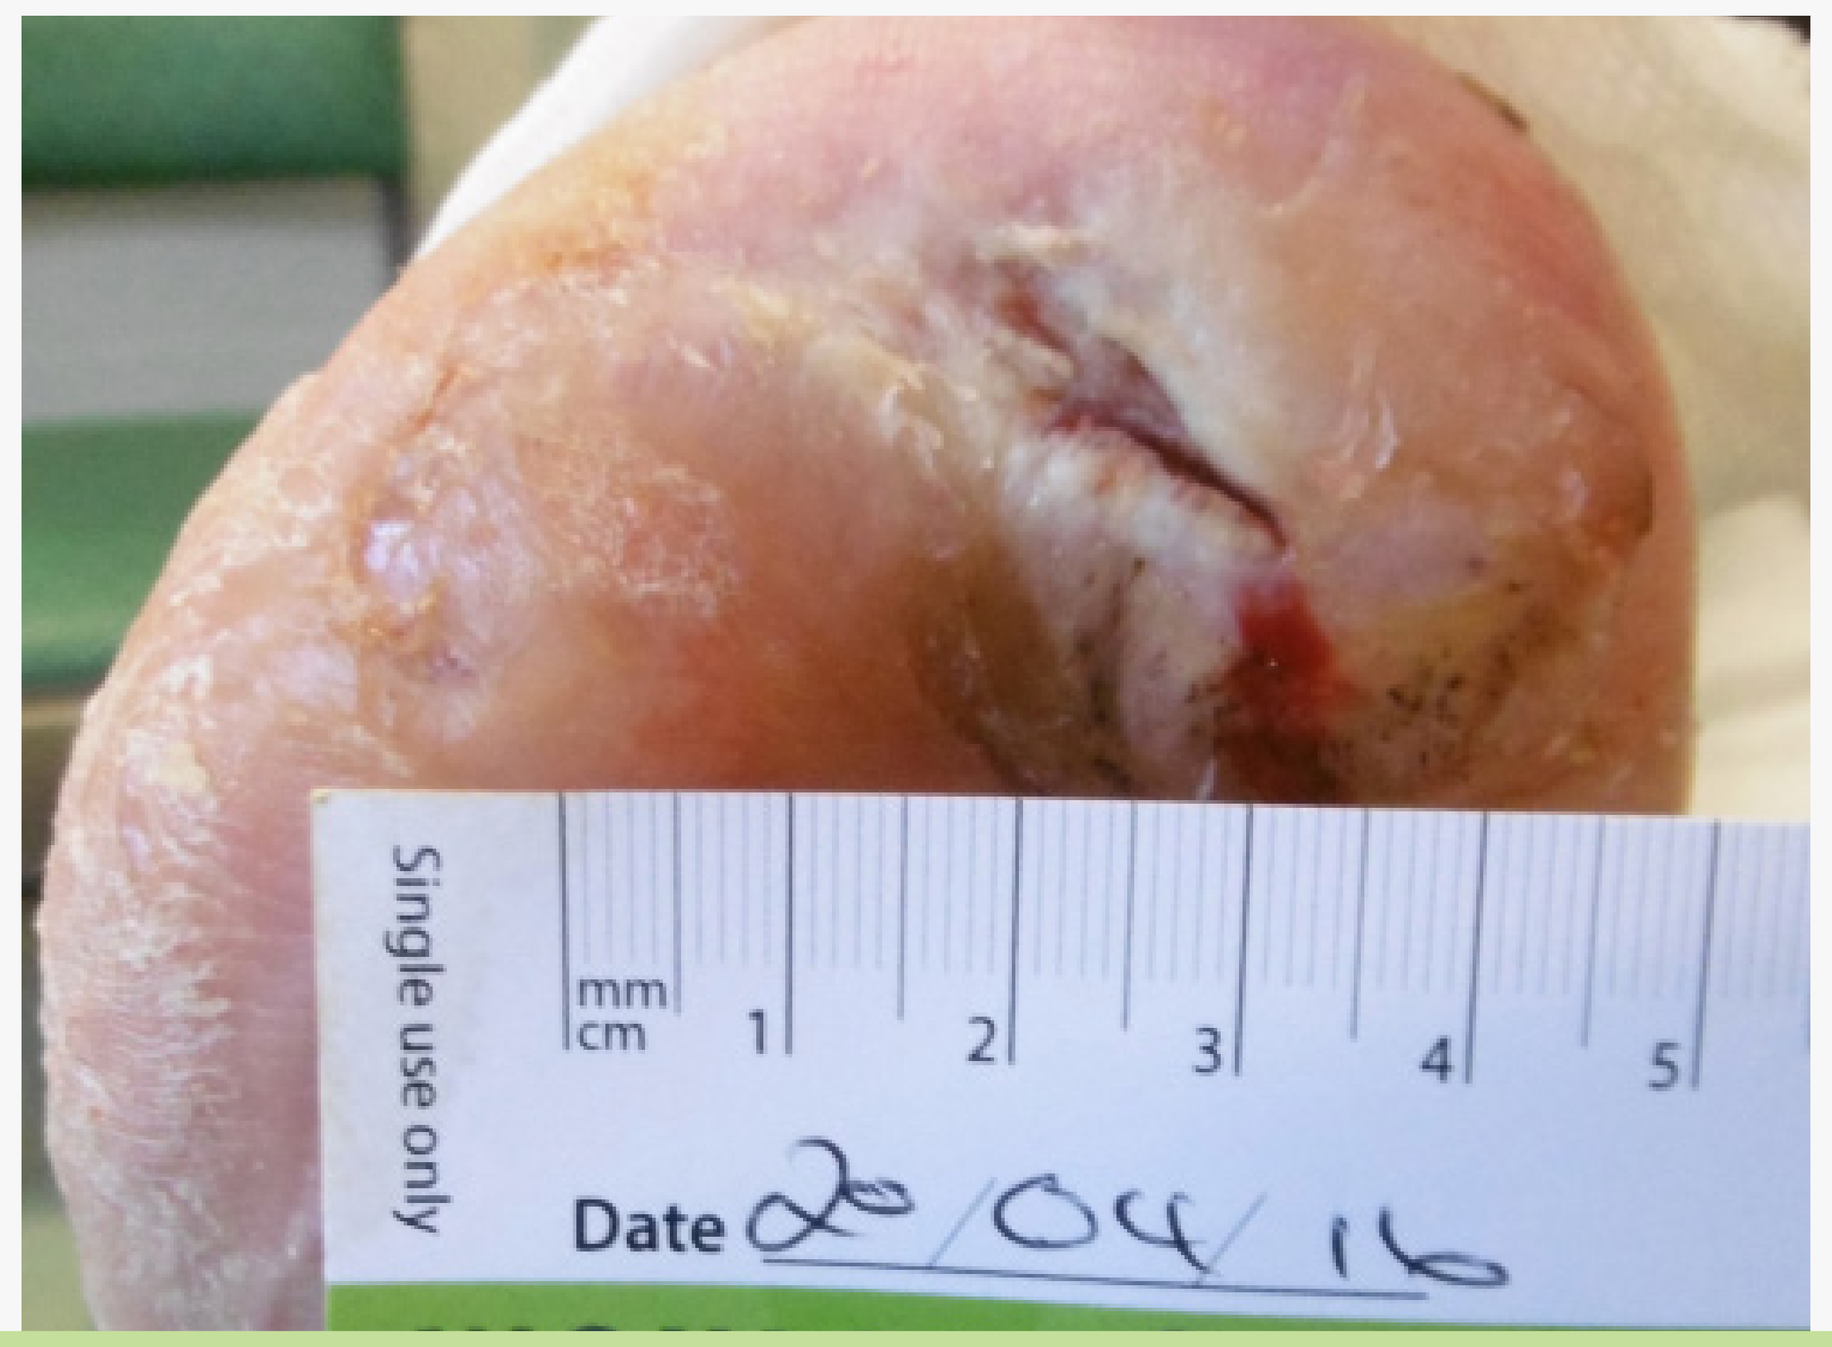
Single use only
mm
cm
1
2
3
4
5
Date 20/04/16

Week 3

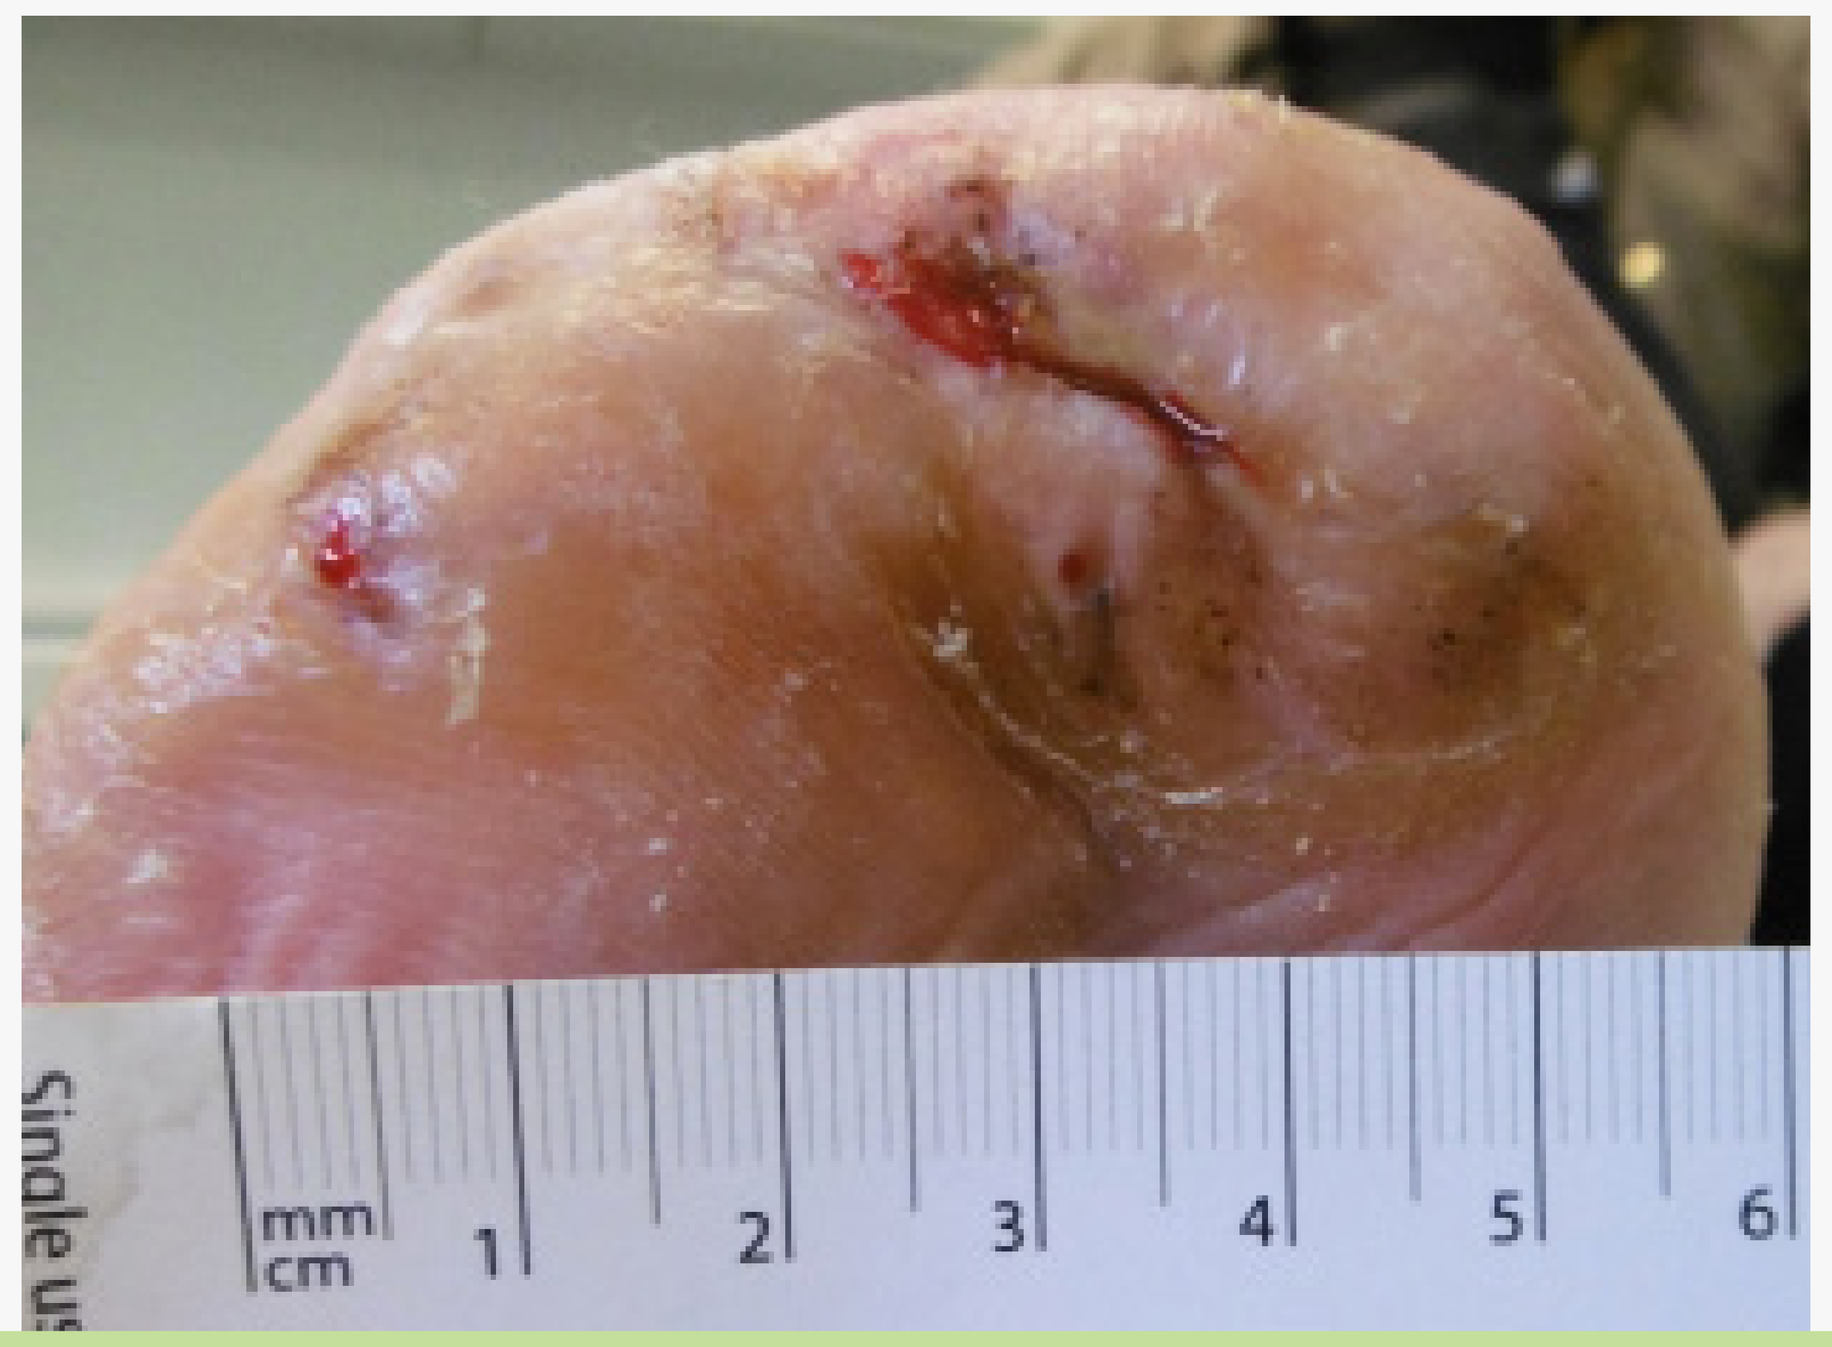
mm
cm
1
2
3
4
5
6

Week 5

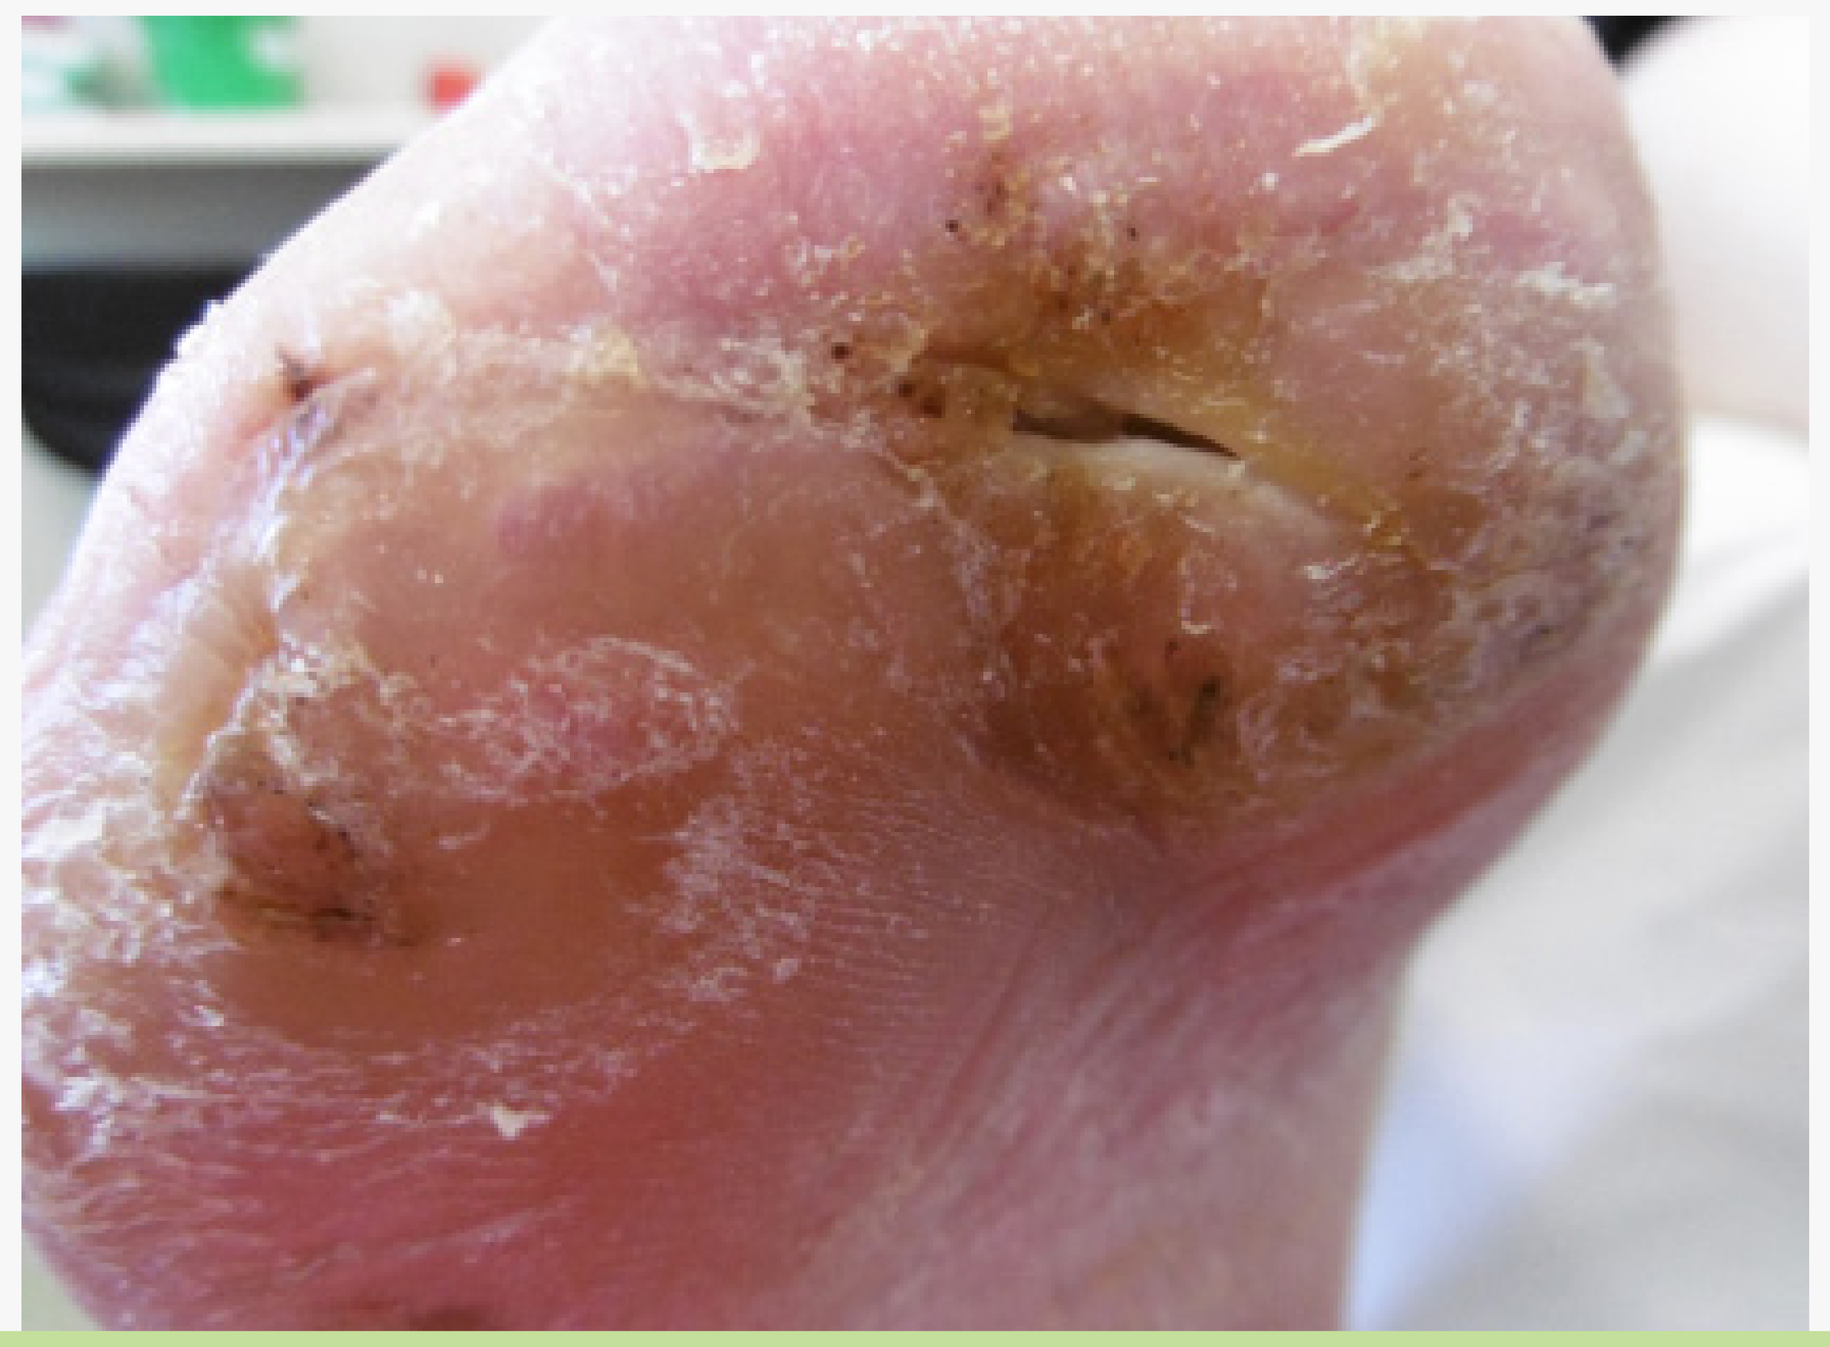

Week 7

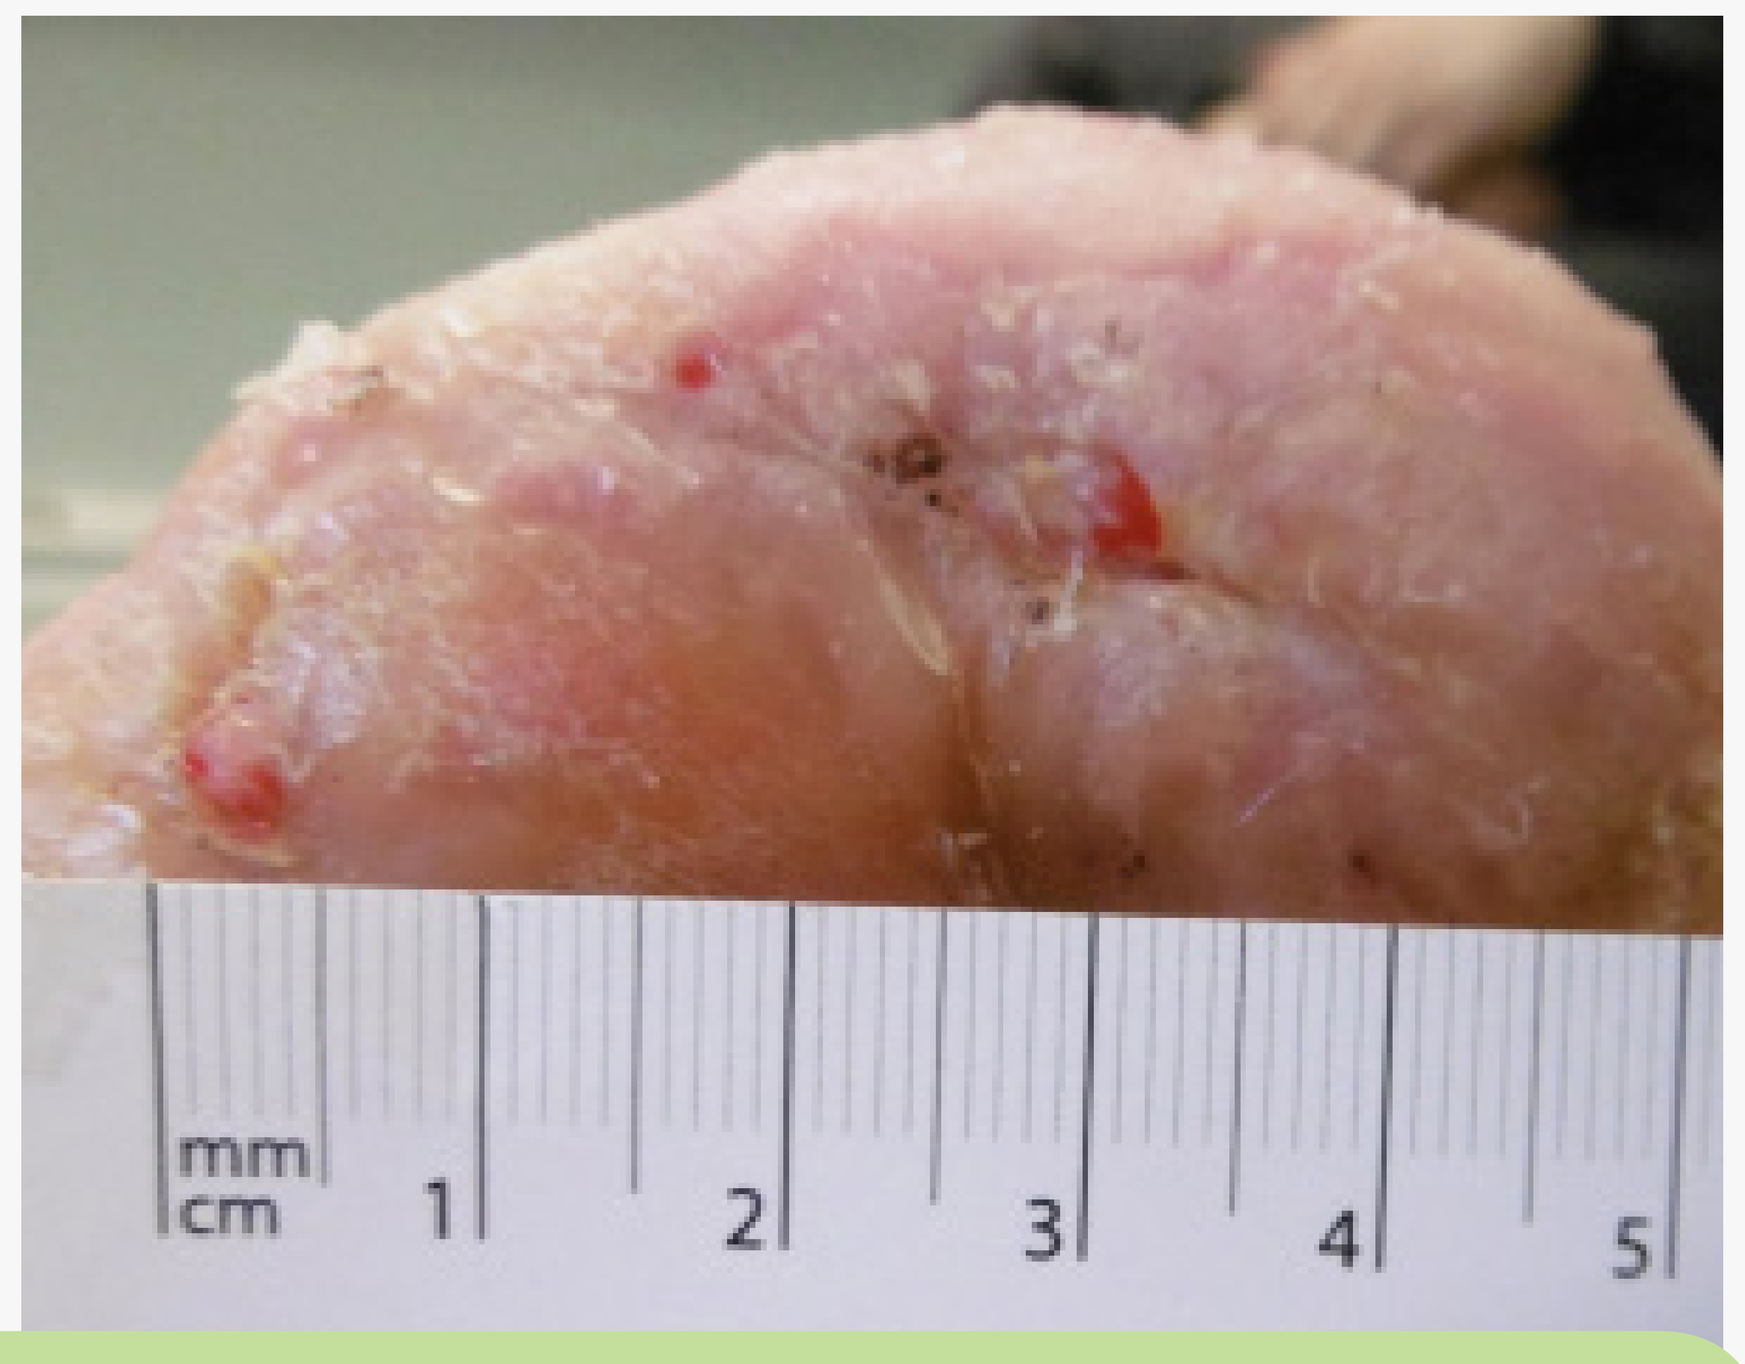
mm
cm
1
2
3
4
5

Week 9

Results:

In both patients the wounds healed and have remained intact since stopping the trial 10 months ago. Comfort on application and removal of dressing were reported as excellent and the pain levels were reduced in case study 1. The gel containing Beta-glucan was reported as very easy to apply.

Conclusion:

The gel with Beta-glucan was found to be an appropriate dressing in the management of 3 non-infected diabetic foot ulcers with low to moderate exudate levels. It appeared to promote healing in wounds that were relatively static. The dressing produced very positive patient outcomes and feedback, and was very easy to apply.

***the gel with Beta-glucan is marketed as Woulgan® Gel**

This poster was supported by an educational grant from Biotec Beta-Glucans, Tromso, Norway